Comparison of Changes in Fat Mass in Participants with PWS Treated with DCCR to Those in the NIH Natural History Study

Jennifer L. Miller MD[1], Daniel J. Driscoll MD, PhD[1], Jack A. Yanovski MD, PhD[2], Neil M. Cowen PhD[3], Kristen Yen[3], Shaila Ballal[3], Anish Bhatnagar MD[3], Merlin G. Butler MD, PhD[4]

[1]University of Florida, Gainesville, FL, USA, [2]*Eunice Kennedy Shriver* National Institute of Child Health and Human Development, Bethesda, MD USA [3]Soleno Therapeutics, Inc., Redwood City, CA, USA, [4]University of Kansas Medical Center, Kansas City, KS, USA.

Abstract #43
United in HOPE
2025 PWS Conference
June 25-26, 2025
Phoenix, AZ

INTRODUCTION

Prader-Willi syndrome (PWS) is a rare neurobehavioral-metabolic genetic disorder characterized by hypotonia, neurocognitive problems, behavioral difficulties, hypogonadism, endocrinopathies, hyperphagia and obesity.

Diazoxide Choline Extended-Release (DCCR) tablets have recently been approved by the FDA as VYKAT™ XR for the treatment of hyperphagia in adults and pediatric patients 4 years of age and older with PWS.[1]

This study was undertaken to compare the fat mass accumulation of participants in the Phase 3 development program for DCCR to that of participants in the NIH funded PWS Natural History Study (NIH NHS).

METHODS

C601 was a placebo-controlled Phase 3 study of DCCR in participants with genetically confirmed PWS, age 4 years and older with hyperphagia.[2] C602 was a long-term, open-label extension to C601.[3] The NIH NHS enrolled PWS participants of any age.[4] Using an independent Contract Research Organization, a cohort from the natural history study were matched for the weight and age criteria of C601 for comparison. The primary endpoint was body fat mass change from Baseline to 2 years. Baseline of the C601/C602 cohort was the last pre-DCCR assessment.

Body fat mass (by DXA) was analyzed by propensity score adjusted ANCOVA. Other endpoints included percent fat mass and fat mass index (FMI). Propensity scores were derived from a logistic regression model adjusting for age, sex, baseline weight (kg), and PWS genetic type (deletion vs. non-deletion).

RESULTS

- The C601/C602 cohort included 115 participants of whom 57 (49.6%) had obesity (based on body mass index) while the NIH NHS cohort included 99 participants among whom 52 (52.5%) had obesity. The demographic and baseline characteristics of the cohorts were comparable with two exceptions (**Table 1**).

Table 1. Baseline Characteristics of Participants in the C601/C602-OLE and NIH NHS Cohorts

	All Subjects		Subjects with Obesity	
	C601/ C602-OLE (N = 115)	NIH NHS (N = 99)	C601/ C602-OLE (N = 57)	NIH NHS (N = 52)
Age (mean [SD]), years	12.9 (6.20)	16.2 (12.57)	13.1 (6.7)	15.0 (11.33)
Sex (% female/male)	57.4/42.6	55.6/44.4	57.9/42.1	53.8/46.2
Weight (mean [SD]), kg	60.1 (29.65)	56.6 (26.93)	73.0 (33.88)	65.5 (29.27)
Body Fat Mass (mean[SD]), kg	26.4 (16.94)	25.7 (16.57)	35.5 (18.48)	33.1 (17.69)
PWS Class (% Del/Non-del)	62.6/37.4	58.6/41.4	61.4/38.6	57.7/42.3
GH Use[a]				
Yes, at both visits	83.5%	53.5%	73.7%	46.2%
No, at both visits	16.5%	35.4%	26.3%	42.3%
At one visit or unknown	0.0%	11.1%	0.0%	11.5%

[a] The cohorts differ primarily because of the period in which they were conducted resulting in differential GH use, the NIH NHS ran from 2008 to 2014 while C601/C602 ran from 2018 to 2023.

Abbreviations: OLE, open-label extension; NIH NHS, NIH funded PWS Natural History Study; PWS, Prader-Willi syndrome; SD, standard deviation; Del, deletion; GH, growth hormone.

- Because of a lack of availability of data in the window or lack of some data needed to calculate a propensity score, only 87 individuals from the C601/C602 cohort were included in the primary analysis. Forty-eight of these individuals had obesity.
- For the analysis of fat mass percent change data was available on 55 individuals in the C601/C602 cohort, 23 of which had obesity. These numbers were reduced to 46 and 18 for the analysis of FMI.
- The primary baseline differences between the cohorts were that the NIH NHS cohort was, on average, about 3 years older (16.2 years vs. 12.9 years) with a lower rate of growth hormone (GH) use (53.5% vs 83.5%). Propensity scores were used to address the differences in the ANCOVA.
- Compared to the NIH NHS DCCR treatment was associated with significantly reduced body fat mass at 2 years among all participants (LS mean difference [95% CI] -4.4 kg [-6.15, -2.63], p<0.001) and in participants with obesity (LS mean difference [95% CI] -7.1 kg [-10.31, -3.98], p<0.001) (**Figure 1**). In all participants and obese participants fat mass loss occurred with DCCR treatment while it increased on placebo.

Figure 1. Fat Mass Change from Baseline to 2 Years (Propensity Adjusted Analyses)

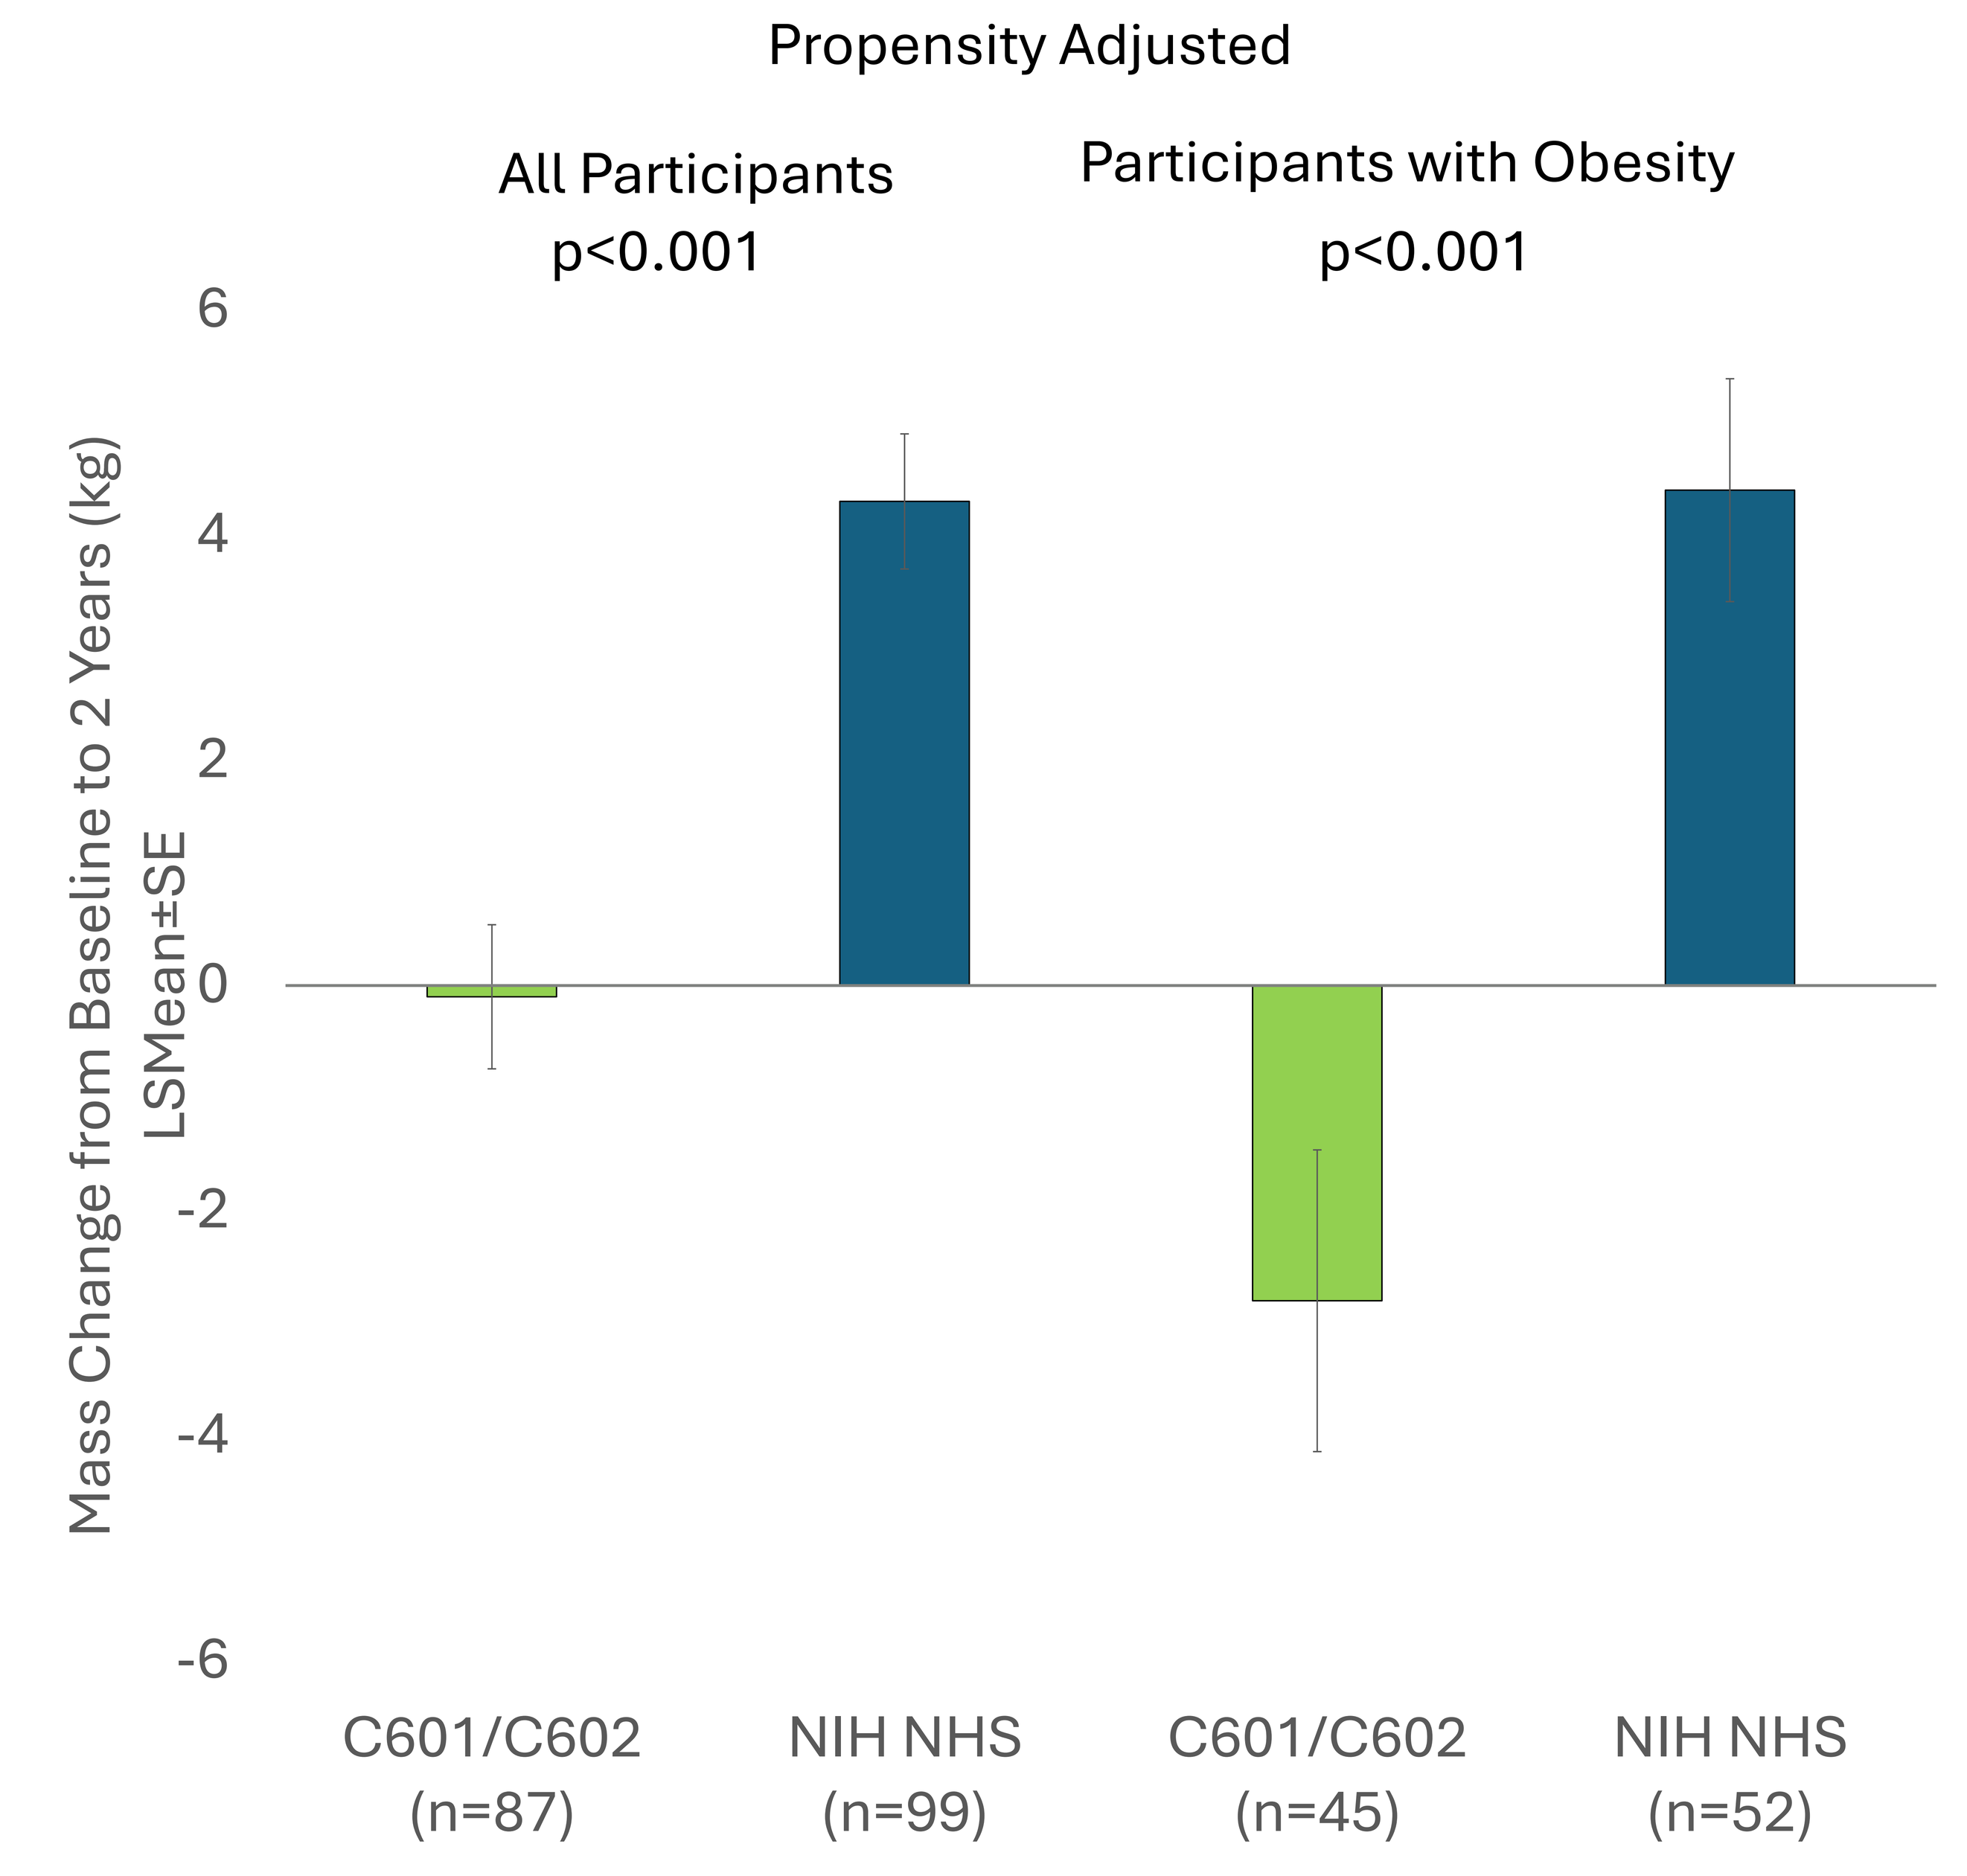
Propensity Adjusted
All Participants
p<0.001
Participants with Obesity
p<0.001
Mass Change from Baseline to 2 Years (kg) LSMean±SE
6
4
2
0
-2
-4
-6
C601/C602 (n=87)
NIH NHS (n=99)
C601/C602 (n=45)
NIH NHS (n=52)

Abbreviations: LSMean, least square means; NIH NHS, NIH funded PWS Natural History Study.

Figure 2. Fat Mass Percent Change from Baseline to 2 Years Using NIH NHS Timespan

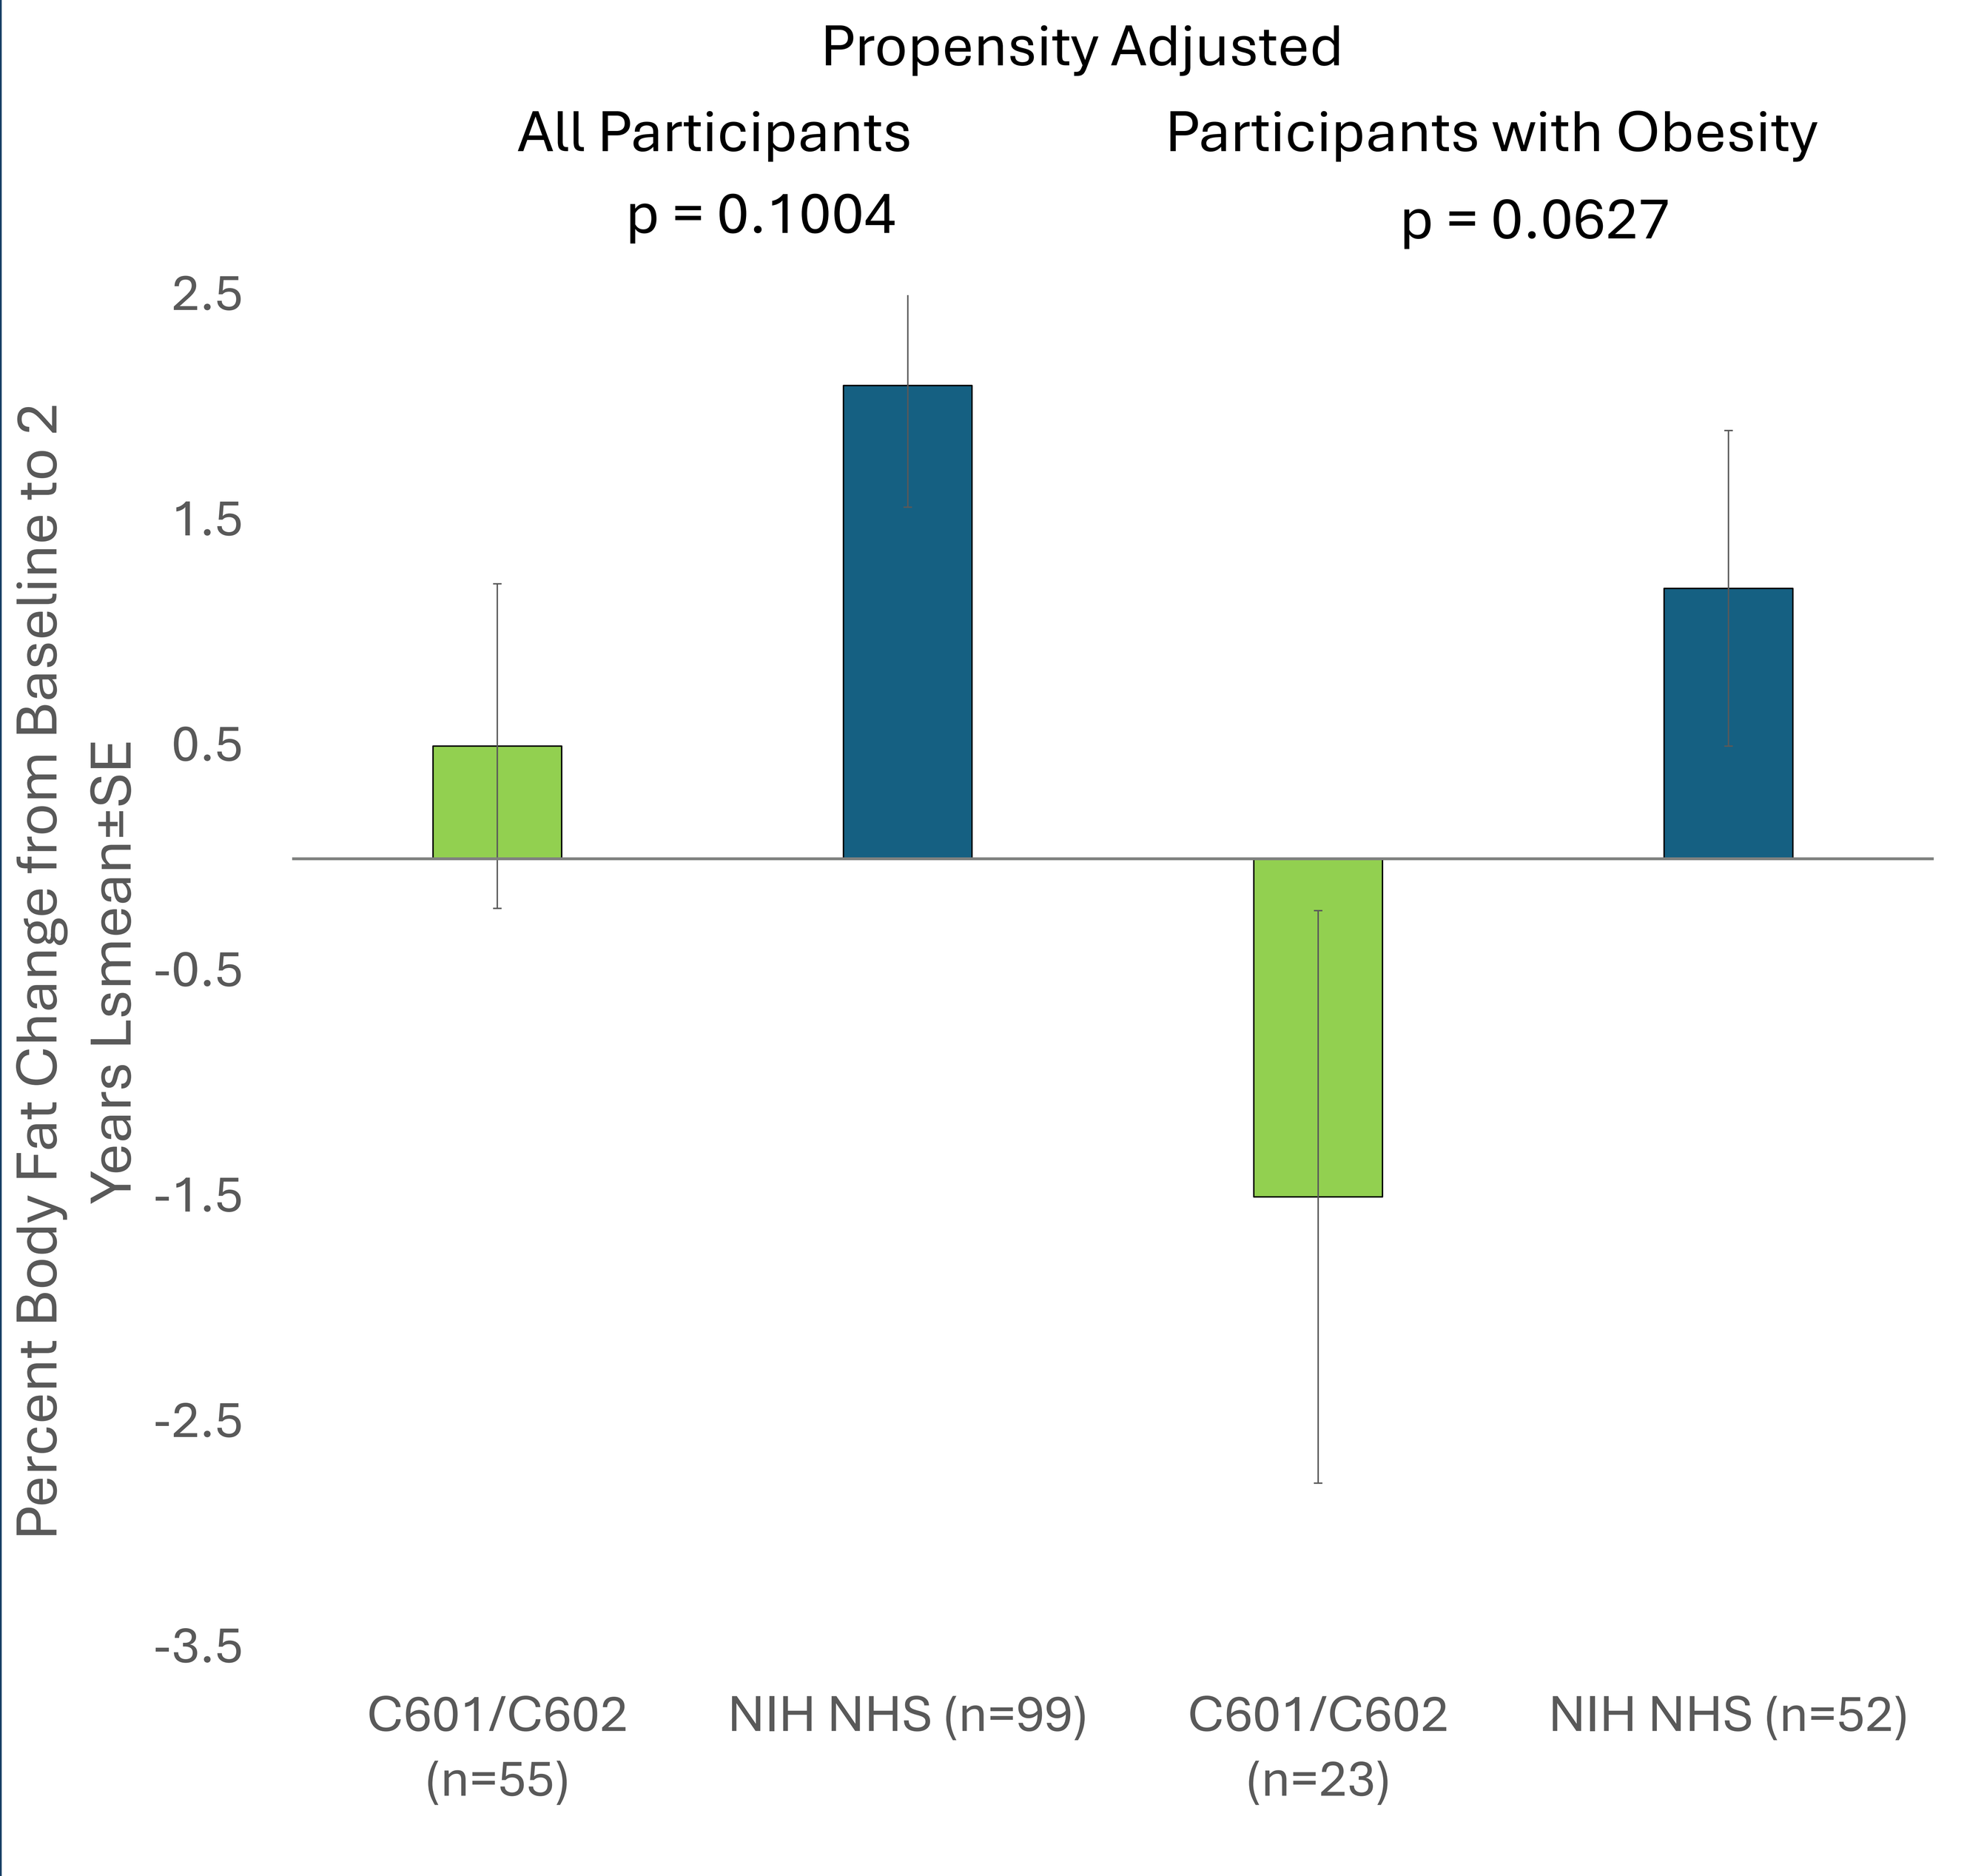
Propensity Adjusted
All Participants
p = 0.1004
Participants with Obesity
p = 0.0627
Percent Body Fat Change from Baseline to 2 Years Lsmean±SE
2.5
1.5
0.5
-0.5
-1.5
-2.5
-3.5
C601/C602 (n=55)
NIH NHS (n=99)
C601/C602 (n=23)
NIH NHS (n=52)

Abbreviations: LSmean, least square means; NIH NHS, NIH funded PWS Natural History Study.

- Compared to the NIH NHS, DCCR treatment was associated with reduced percent body fat at 2 years that approached but did not achieve significance among all participants and in participants with obesity (**Figure 2**).
- FMI change comparison was not significant among all participants (LS mean [95% CI] difference -0.6 [-1.53, 0.38], p=0.2364) or among participants with obesity (LS mean [95% CI] difference -0.8 [-2.52, 0.89], p=0.3405). These results were likely impacted by the small sample size for this analysis.

CONCLUSIONS

People with PWS are prone to the accumulation of excess body fat. DCCR treatment reduces the accumulation of body fat in people with PWS relative to the natural history of the syndrome. These changes in body fat along with the previously reported improvements in hyperphagia, lean body mass and behavioral responses to DCCR treatment are likely to contribute to improved quality of life for patients with PWS.

ACKNOWLEDGMENTS

Soleno Sponsored Clinical Studies C601 and C602, NIH NHS was conducted by NIH funded Rare Disease Clinical Research Network, Grant U54 RR019478.

REFERENCES

1. VYKAT™ XR [package insert]. Soleno Therapeutics, Inc. 2025.
2. Miller J, et al. *J Clin Endocrinol Metab*. 2023; 108:1676-1685.
3. Miller J, et al. *Obesity (Silver Spring)*. 2024; 32(2):252-261.
4. Butler MG, et al. *Am J Med Genet A*. 2018; 176(2):368-375.

CONTACT INFORMATION

First author: millejl@peds.ufl.edu; presenting author: neil@soleno.life; senior author: mbutler4@kumc.edu.

ELECTRONIC POSTER
Copies obtained through the QR code are for personal use only.

Soleno Therapeutics, Inc. © 2025